Development of a Multi-biomarker Risk Score based on Serum Proteins by the Prognostic Lung Fibrosis Consortium (PROLIFIC)

Peter Schafer[1*,13], John Rassa[1], Agnes Seyda[1], Sarah Hersey[1], Troy Tremaine[2], Dominic Eisinger[2*], Jenna Melin[2], Robert Bencher[2*], Margaret Neighbors[3], Majd Mouded[4*], Scott Turner[5*], Martin Decaris[5], Yasmina Bauer[6*], Cheryl Nickerson-Nutter[7], Dana Ball[7*], Jeffrey Larson[8], James Swaney[9], Sharlene R. Lim[10], Andrew Whiteman[10], Alison MacKinnon[11], Francesco Marabita[12], Jennifer Mefford[13], Scott Staszak[13], Adam Martin-Schwarze[14], Zheshi Zheng[14], Peter X. Song[14], Joseph A. Lasky[15]

[1]Bristol Myers Squibb, Princeton, NJ, USA, [2]Rules-Based Medicine, an IQVIA business, Austin, TX, USA, [3]Genentech Inc., South San Francisco, CA, USA, [4]Novartis Institutes for Biomedical Research, Inc. Boston, MA, USA, [5]Pliant Therapeutics, South San Francisco, CA, USA, [6]Galapagos NV, Basel, Switzerland, [7]Three Lakes Foundation Trust, Northbrook, IL, USA, [8]Tvardi Therapeutics, Houston, TX, USA, [9]Lassen Therapeutics, San Diego, CA, USA, [10]Gilead Sciences, Foster City, CA, USA, [11]Galecto Biotech, Boston, MA, USA, [12]Chiesi Corporate R&D, Solna, Sweden, [13]Pulmonary Fibrosis Foundation, Chicago, IL, USA, [14]Tulane University School of Medicine, New Orleans, LA, USA, [15]Department of Biostatistics, University of Michigan, Ann Arbor, MI, USA . * Affiliation when the work was conducted

Introduction

Despite >20 years of IPF biomarker discovery, there are no approved prognostic biomarker tests available for patient stratification or eligibility as a drug development tool. To address this unmet need, the Prognostic Lung Fibrosis Consortium (PROLIFIC) was formed to develop well-qualified biomarker assays, which were applied to large IPF patient serum collections to derive and validate the PROLIFIC Prognostic Risk Score.

Assay Development

Twelve IPF biomarkers were selected from previous literature and immunoassays were developed at Rules-Based Medicine in either in single-plex or multiplex format utilized the Luminex® xMAP® platform under design control. Qualification studies were conducted to meet pre-defined performance characteristics with multiple lots of reagents.

Serum protein	LLOQ	ULOQ	Accuracy (% diff)	Inter-Assay Precision (CV)	Freeze-Thaw Stability (% recovery up to 5x)	Matrix Interference (% recovery)	Parallelism (% recovery)	Sample Repro-ducibility (%CV)	Short-term Analyte Stability (% variability)
ICAM-1 (ng/mL)	32	17300	-2 to 1%	6-12%	90-105%	99%	113%	<10%	<10%
KL-6 (IU/mL)	1·7	2450	-2 to 3%	7-10%	80-102%	94%	97%	<10%	<10%
PAI-1 (ng/mL)	1·5	1680	-2 to 1%	7-8%	93-104%	111%	104%	<10%	<10%
PARC (CCL18) (ng/mL)	0·85	1730	-2 to 1%	5-7%	94-111%	101%	107%	<10%	<10%
SP-D (ng/mL)	11	15000	-2 to 1%	8-12%	86-108%	108%	112%	<10%	<10%
TN-C (ng/mL)	154	228000	-3 to 0%	5-11%	85-102%	113%	94%	<10%	<10%
CA-125 (IU/mL)	9·7	8150	-1 to 6%	9-13%	80-108%	112%	115%	<20%	<10%
CYFRA 21-1 (ng/mL)	0·51	326	3 to 6%	9-11%	80-100%	103%	106%	<15%	<15%
BLC (CXCL13) (pg/mL)	14	18000	0%	10-12%	85-100%	111%	93%	<15%	<15%
Periostin* (ng/mL)	18	18500	0%	11-20%	90-100%	107%	90%	<10%	<10%
MMP-7 (ng/mL)	0·050	153	-1 to 0%	9-13%	86-105%	100%	114%	<10%	<10%
CA-19-9 (IU/mL)	2·1	2940	-7 to -2%	6-9%	90-100%	110%	117%	<10%	<10%

*Periostin had unsatisfactory proficiency testing outside the range for CLIA specifications and has been removed.

Baseline sera from IPF patients in the PFF Patient Registry (N=657) were used for statistical analyses to define a composite binary outcome of death, lung transplant, or ≥ 10% relative decline in % predicted FVC in one year. LASSO penalized logistic regression was used to select the top biomarkers: SP-D, CXCL13, TNC, and MMP-7 (Figure 1).

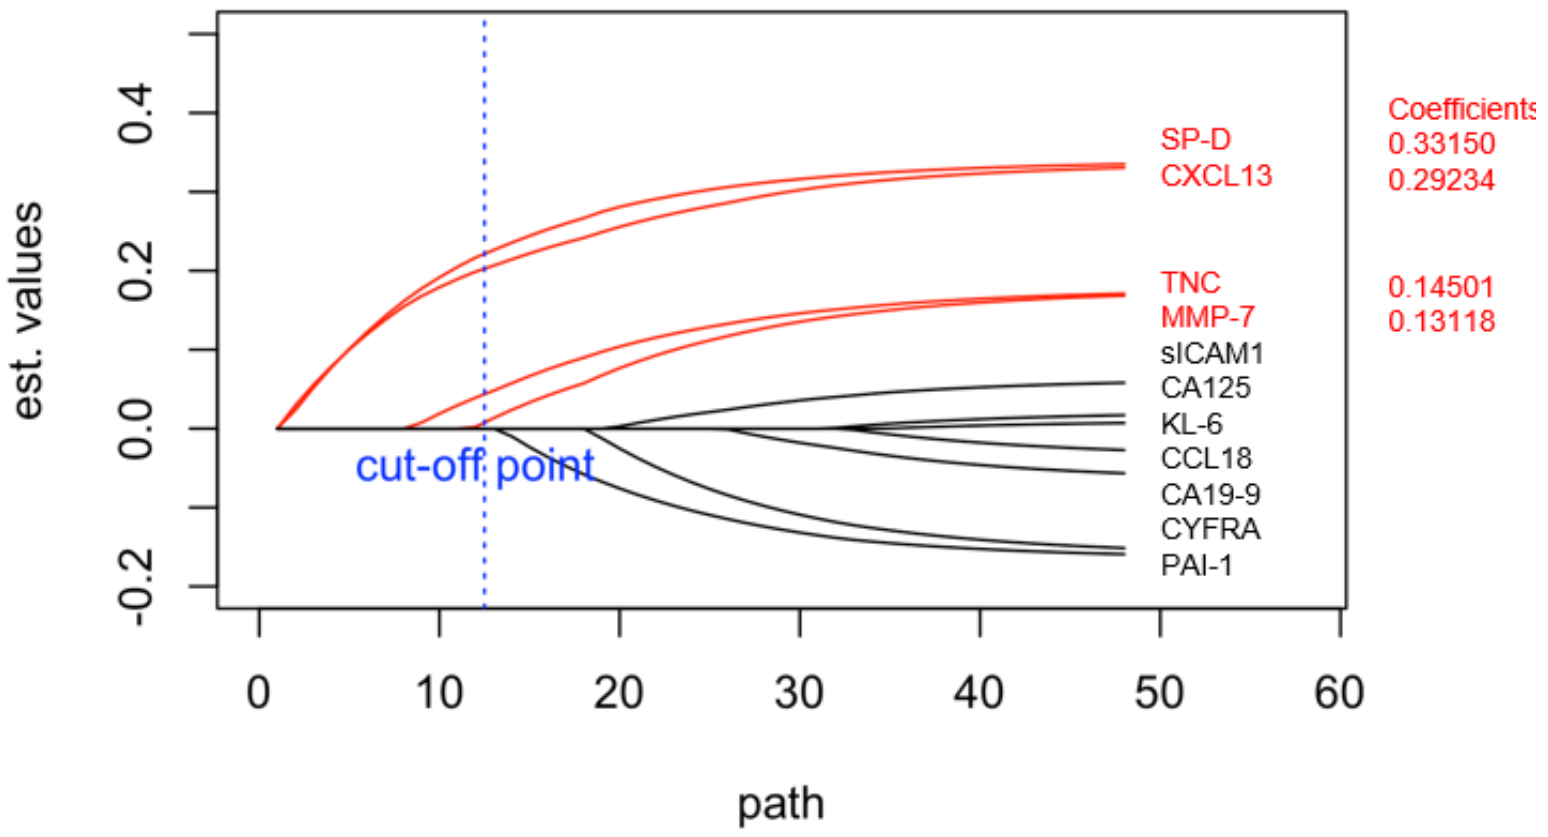
est. values
0.4
0.2
0.0
-0.2
path
0
10
20
30
40
50
60
cut-off point
SP-D
CXCL13
TNC
MMP-7
sICAM1
CA125
KL-6
CCL18
CA19-9
CYFRA
PAI-1
Coefficients
0.33150
0.29234
0.14501
0.13118

Model Derivation

The selected biomarkers (SP-D, CXCL13, TNC, and MMP-7) were used to fit a logistic model while adjusting for sex, age, BMI, anti-fibrotic medication, smoking packs per year, % predicted FVC, and % predicted DLCO, to derive a risk score:

PROLIFIC Prognostic Risk Score=

$$A + B\ \ln(C_{SPD}) + C\ \ln(C_{CXCL13}) + D\ \ln(C_{TNC}) + E\ \ln(C_{MMP7})$$

The PROLIFIC Prognostic Risk Score was evaluated by Area Under the Receiver Operator Characteristic (AUROC) curve for its predictive performance, applying the optimal cut-off value based on the Youden J statistic: AUC=0.796, sensitivity=0.752, specificity=0.699 (Figure 2).

Model Validation

The PROLIFIC Prognostic Risk Score was validated using an independent dataset comprised of subjects from the placebo control arms of the ISABELA phase 3 studies of ziritaxestat (N=229) applying the same cut-off value: AUC=0.825, sensitivity=0.764, specificity=0.715 (Figure 2).

The PROLIFIC consortium has submitted a Letter of Intent to the FDA Center for Drug Evaluation and Research (CDER) to qualify the PROLIFIC Prognostic Risk Score as a drug development tool under the Biomarker Qualification Program (BQP). The qualified biomarkers may be used as stratification or eligibility criteria in clinical trials and have the potential to provide valuable information that may reduce uncertainty in decisions made during drug development.

Figure 2: Performance of the PROLIFIC Prognostic Risk Score in the derivation and validation datasets.

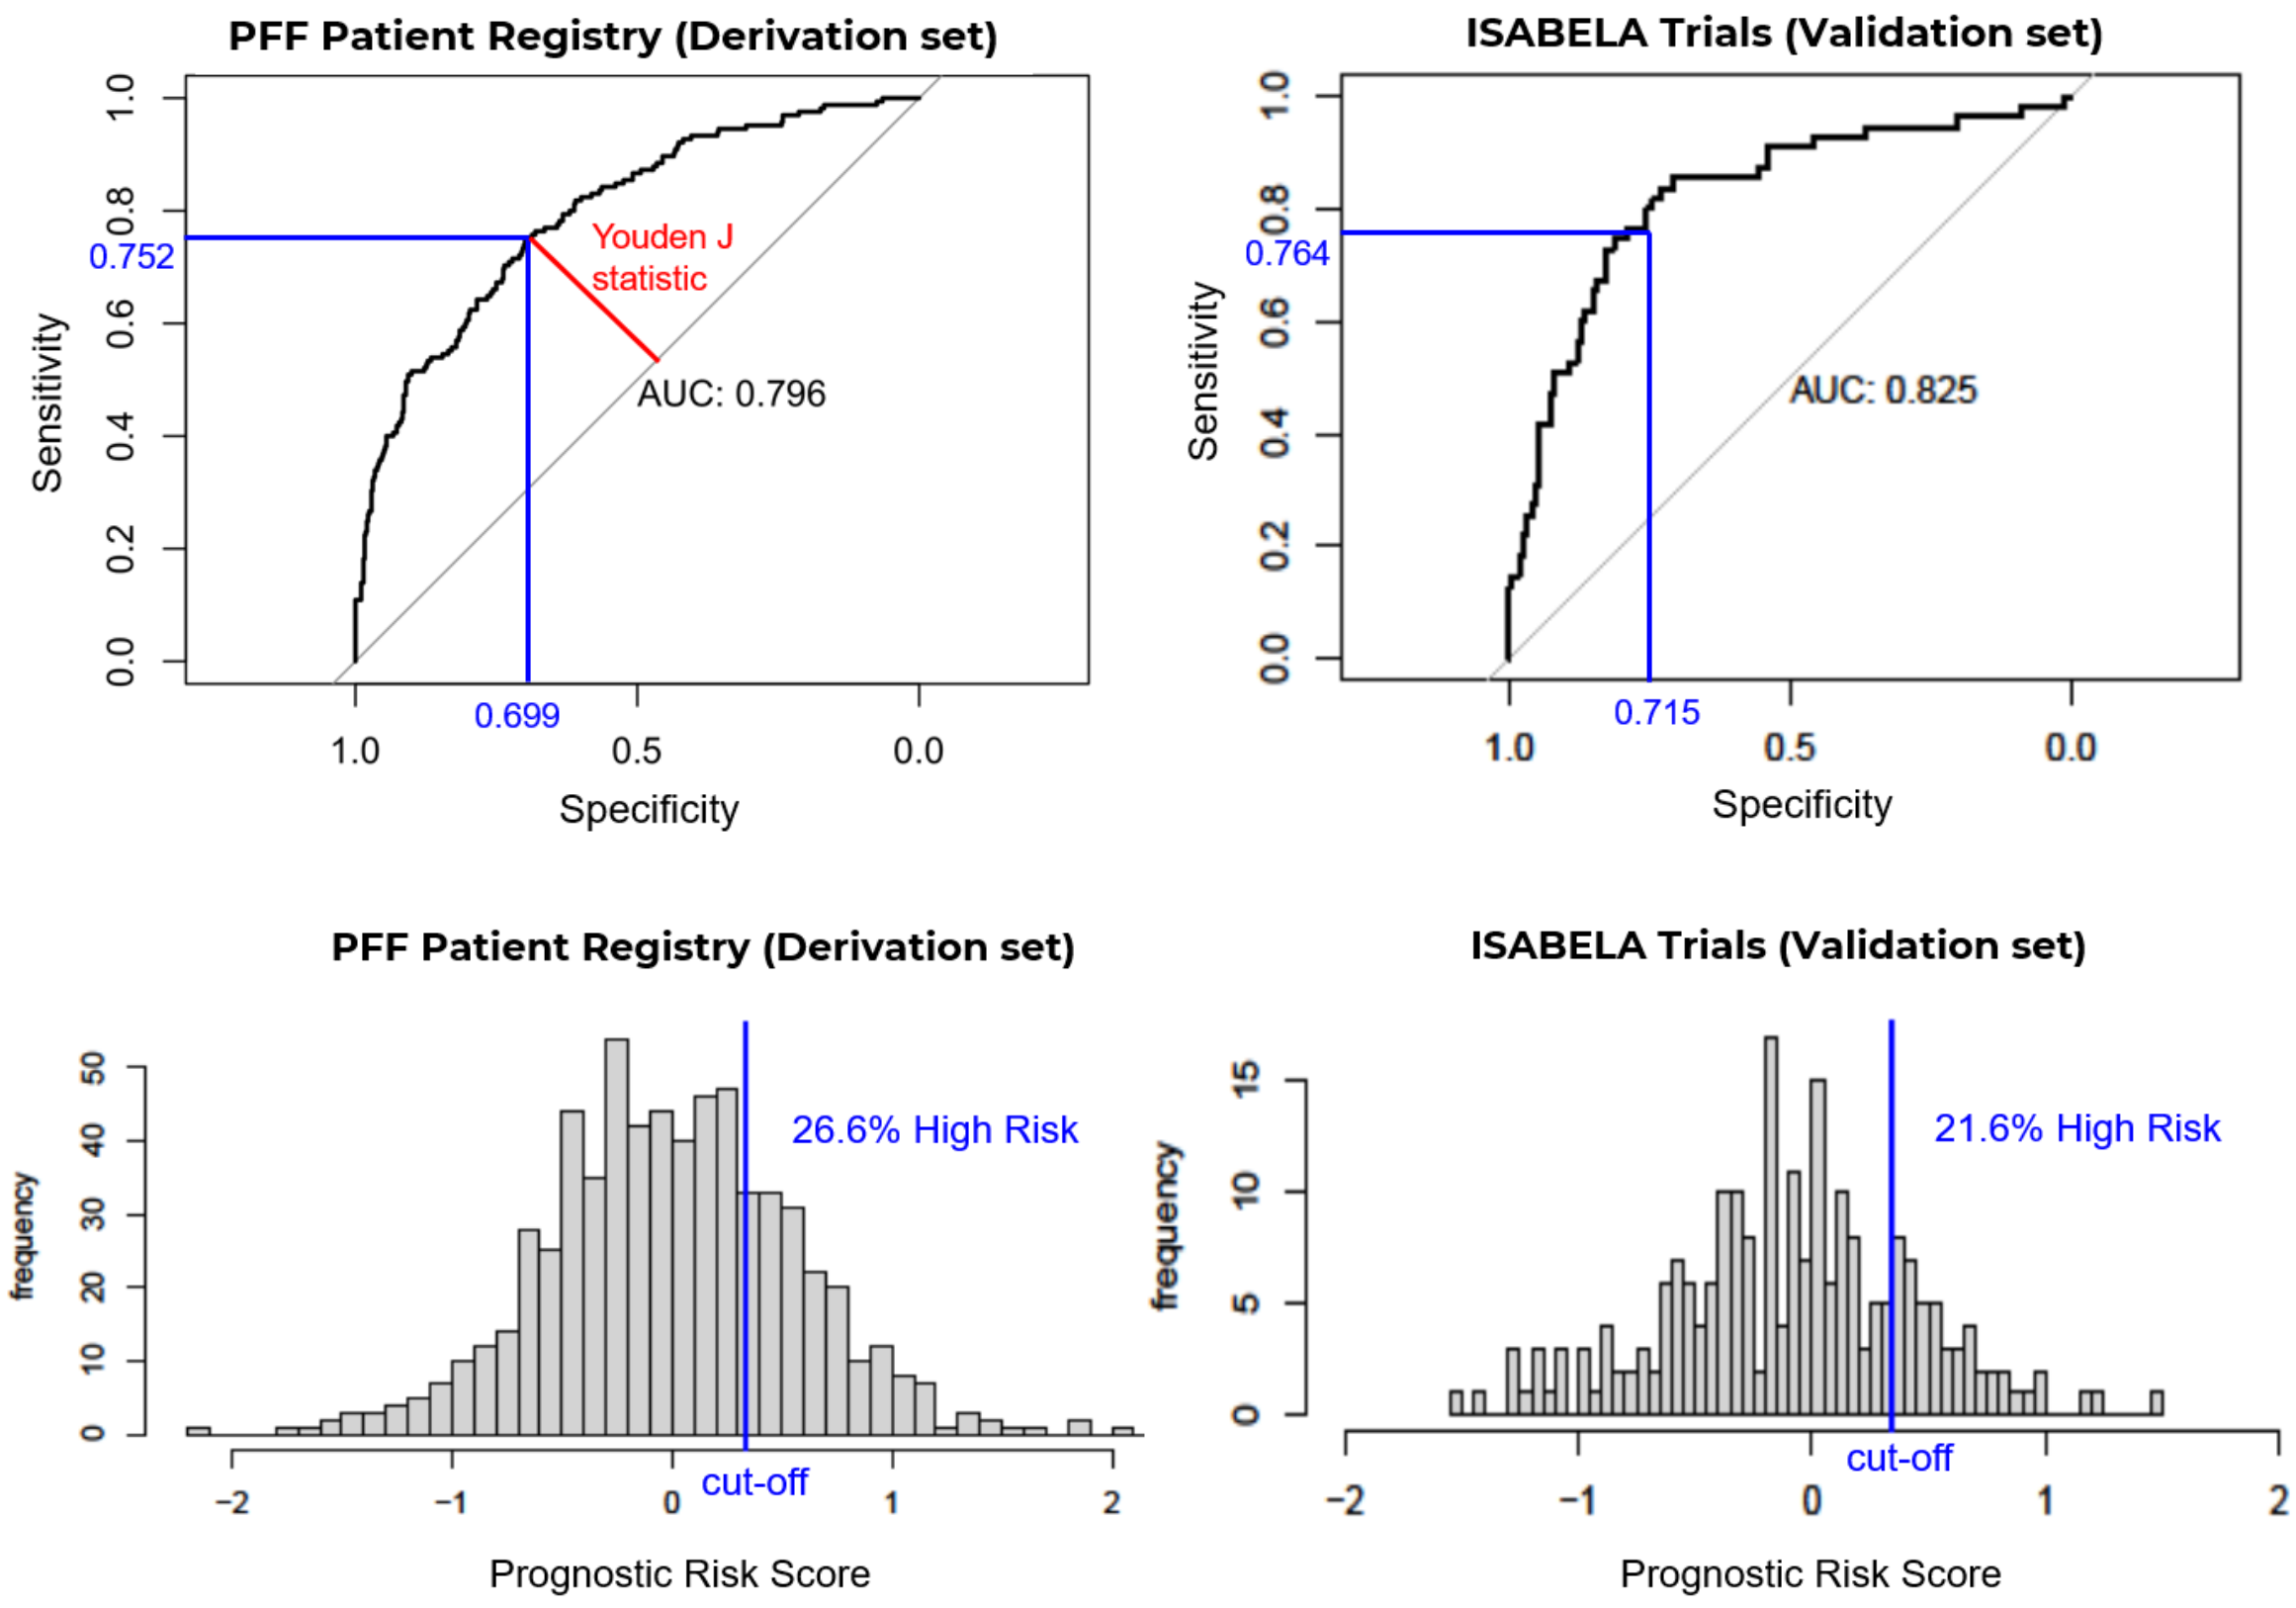
PFF Patient Registry (Derivation set)
Sensitivity
0.752
Youden J
statistic
AUC: 0.796
0.699
Specificity
ISABELA Trials (Validation set)
0.764
AUC: 0.825
0.715
PFF Patient Registry (Derivation set)
frequency
26.6% High Risk
cut-off
Prognostic Risk Score
ISABELA Trials (Validation set)
21.6% High Risk
cut-off
Prognostic Risk Score

Scan to see the IPF assays utilized: